Safety and Pharmacokinetics of MK-8527 Oral Once-monthly: a Phase 2 Study in Adults at Low Risk of HIV-1 Exposure

Kenneth Mayer[1,2]; Philip Kotze[3]; Johannes Lombaard[4]; Yoseph Caraco[5]; Avivit Peer[6]; Nicole Poovan[7]; Satkirin Khalsa[8]; Shishir Khetan[9]; Adrienne E. Shapiro[10]; Mohammed Rassool[11]; Antonio Gonzalez[12]; Yashna Singh[13]; Gary Sinclair[14]; Sharon Riddler[15]; Susan Buchbinder[16]; Michelle Vesay[17]; Brenda Homony[17]; Barbara Evans[17]; Anjana Grandhi[17]; Chenguang Zhang[17]; Maja Skataric[17]; Michael N. Robertson[17]; Yash Kapoor[17]; Randolph P. Matthews[17]; Rebeca M. Plank[17]; Boris Renjifo[17]

[1]Fenway Health and Department of Medicine, Beth Israel Deaconess Medical Center, Boston, MA, USA; [2]Harvard Medical School, Boston, MA, USA; [3]Qhakaza Mbokodo Research Clinic, Ladysmith, South Africa; [4]Josha Research, Bloemfontein, South Africa; [5]Hadassah-Hebrew University Medical Center, Jerusalem, Israel; [6]Rambam Health Care Campus, Haifa, Israel; [7]Wits RHI, University of the Witwatersrand, Johannesburg, South Africa; [8]Albuquerque Clinical Trials, Inc., Albuquerque, NM, USA; [9]Velocity Clinical Research, Rockville, MD, USA; [10]University of Washington & Fred Hutchinson Cancer Center - Seattle HIV Vaccine Trials Unit, Seattle, WA, USA; [11]Helen Joseph Hospital-Clinical HIV Research Unit, Johannesburg, South Africa; [12]Advanced Research for Health Improvement, Immokalee, FL, USA; [13]Desmond Tutu HIV Centre, Institute of Infectious Diseases and Molecular Medicine, University of Cape Town, South Africa; [14]Prism Health North Texas, Oak Cliff Health Center, Dallas, TX, USA; [15]University of Pittsburgh, Pittsburgh, PA, USA; [16]Bridge HIV, San Francisco Department of Public Health, San Francisco, CA, USA; [17]Merck & Co., Inc., Rahway, NJ, USA.

Background

- Despite the approval of several agents for pre-exposure prophylaxis (PrEP), approximately 1.3 million (1.0-1.7 million) people acquired HIV in 2023[1]
- The efficacy of emtricitabine/tenofovir disoproxil fumarate (FTC/TDF) and emtricitabine/tenofovir alafenamide (FTC/TAF) for PrEP is strongly correlated with adherence to daily oral therapy,[2] which can be a challenge for some people who could benefit from PrEP[3]
 - Long-acting oral options for HIV-1 PrEP, with less frequent dosing, are needed
- Once-monthly (QM) oral PrEP was the preferred modality among respondents in a recent online survey[4]
- MK-8527 is a novel, oral, nucleoside reverse transcriptase translocation inhibitor with pharmacokinetic properties supporting QM dosing[5]
- We studied the safety, tolerability, and pharmacokinetics of MK-8527 oral QM in adults with low likelihood of HIV-1 exposure in a randomized, double-blind, placebo-controlled phase 2 clinical study (MK-8527-007, NCT06045507)

MK-8527-007 study design

- Participants were randomized (2:2:2:1) to receive oral monthly doses of MK-8527 (3, 6, or 12 mg) or placebo for up to 6 consecutive months (1 dose every 4 weeks) from day 1 to week 20
- Randomization was stratified by sex (female, male) and region (Africa, non-Africa)
- The study was conducted in Israel, South Africa, and the United States

Figure 1. Phase 2 randomized double-blind, dose-ranging study

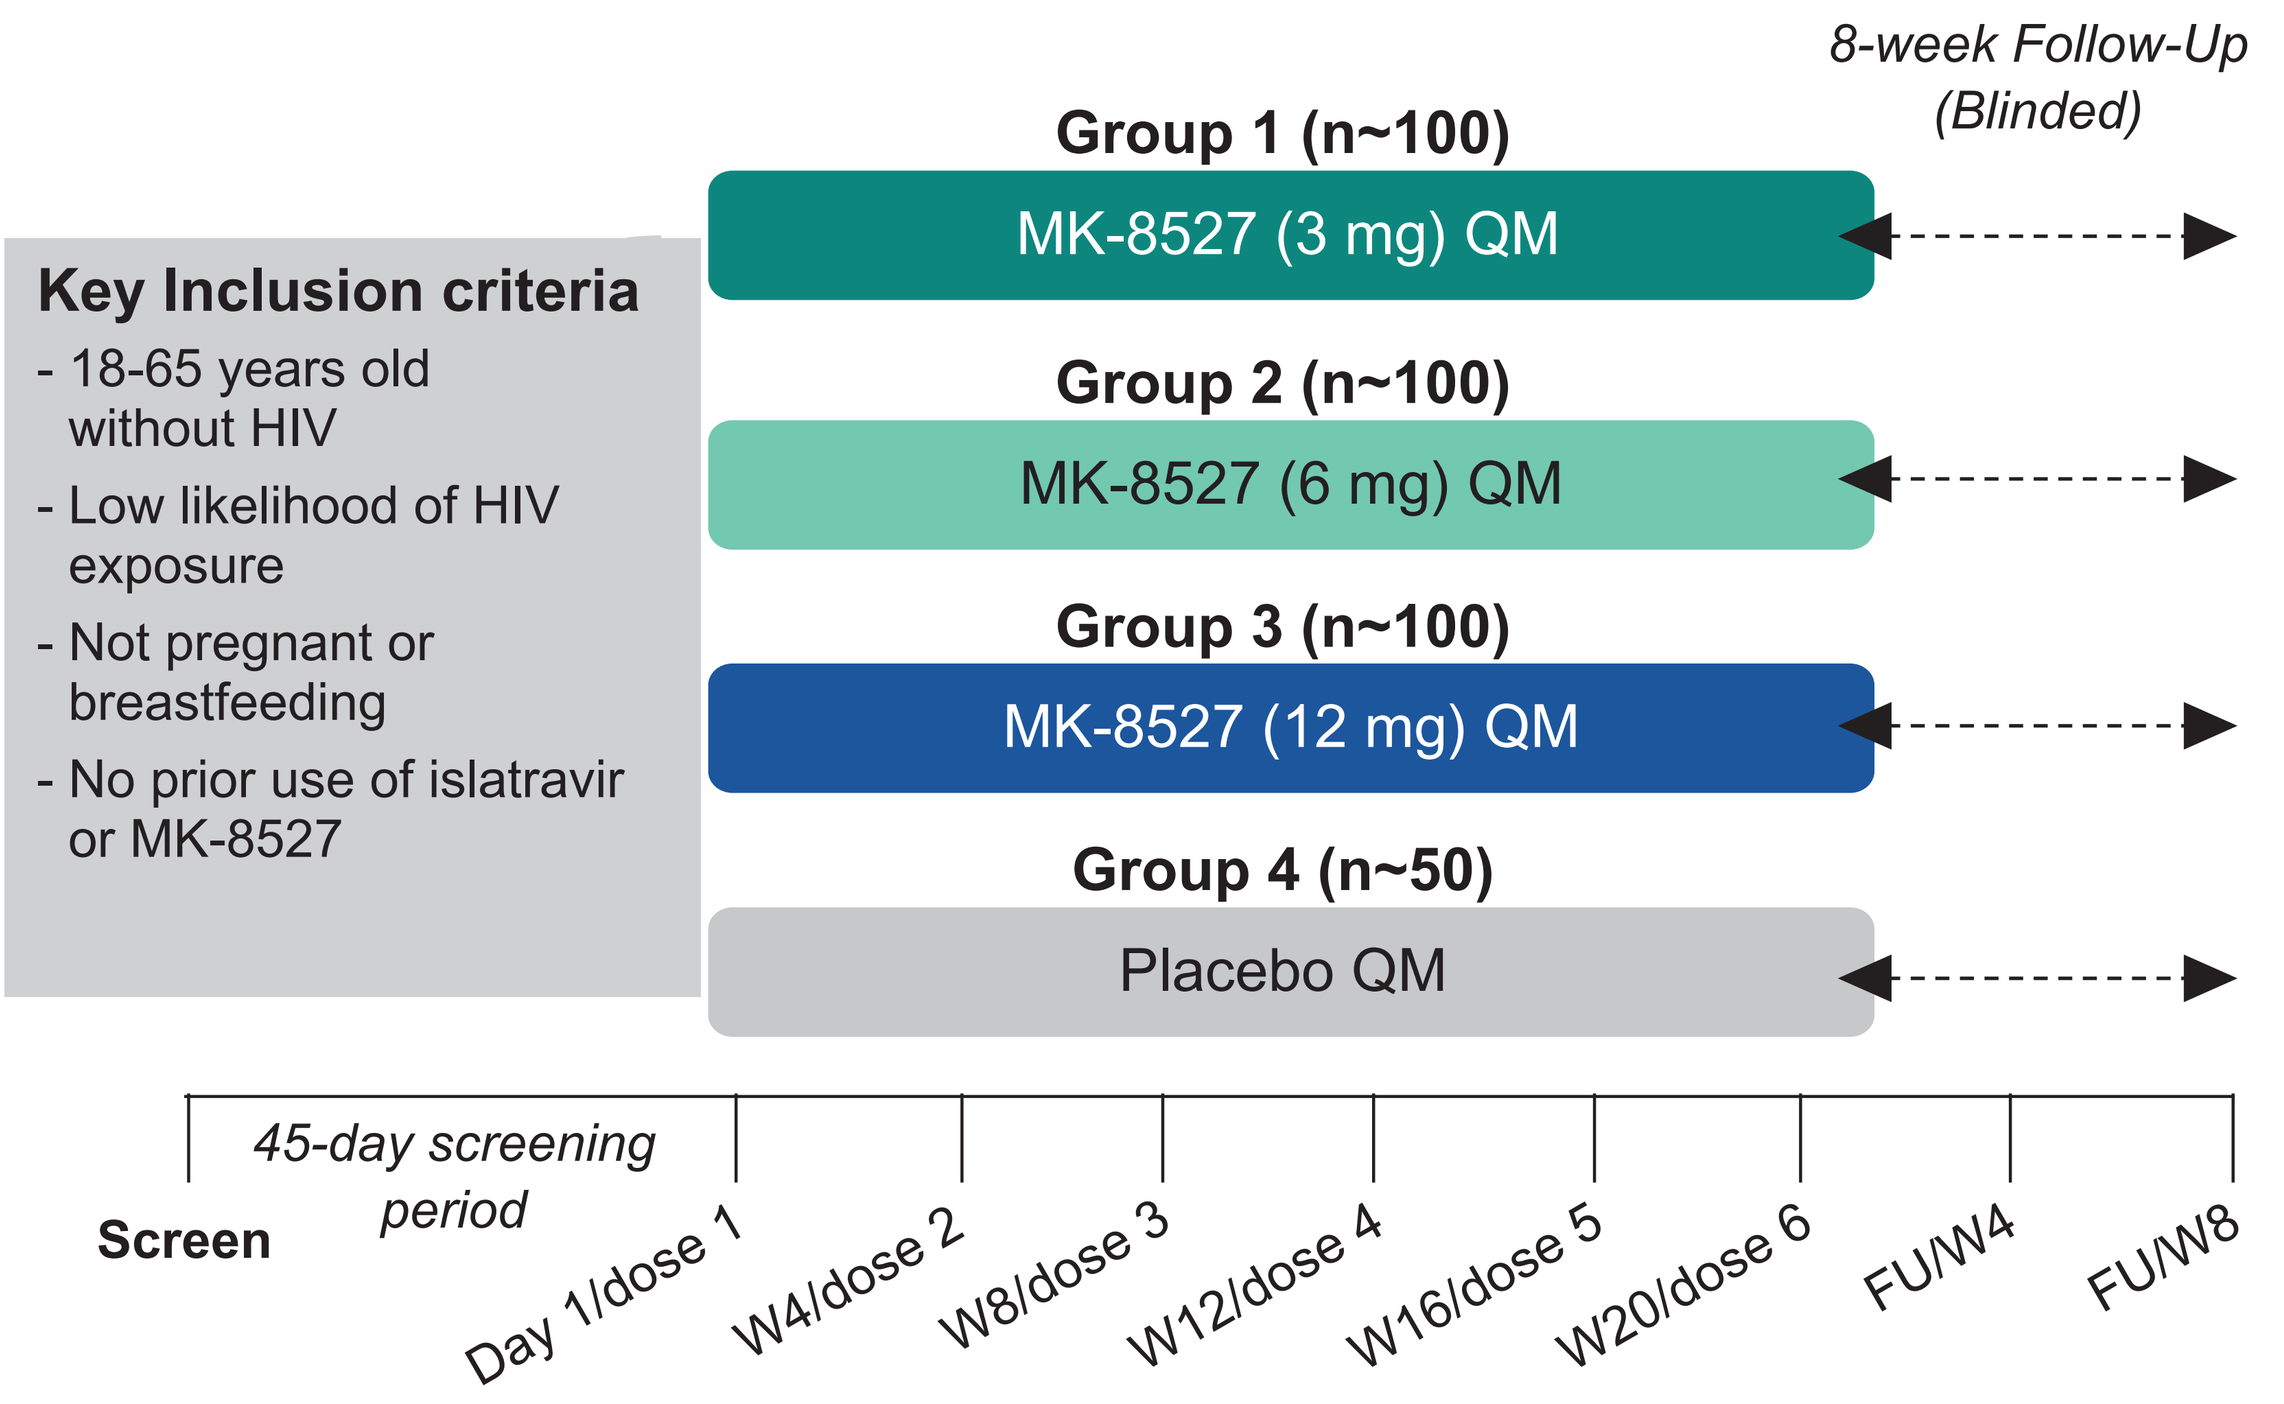
8-week Follow-Up (Blinded)
Group 1 (n~100)
MK-8527 (3 mg) QM
Group 2 (n~100)
MK-8527 (6 mg) QM
Group 3 (n~100)
MK-8527 (12 mg) QM
Group 4 (n~50)
Placebo QM
Key Inclusion criteria
- 18-65 years old without HIV
- Low likelihood of HIV exposure
- Not pregnant or breastfeeding
- No prior use of islatravir or MK-8527
Screen
45-day screening period
Day 1/dose 1
W4/dose 2
W8/dose 3
W12/dose 4
W16/dose 5
W20/dose 6
FU/W4
FU/W8

Assessments

Safety & Tolerability

- Review of adverse events and laboratory tests were performed at each study visit through 8 weeks after last dose
- Study intervention was discontinued for on-treatment decreases (confirmed at 2 consecutive visits) in total lymphocyte count and/or CD4 count, specified as <1.0 x 10^9/L and <500 cells/mm^3, respectively, with a ≥30% decline from baseline
- HIV testing was performed at screening, day 1, and every 4 weeks thereafter

Pharmacokinetics

- MK-8527 in plasma measured in all participants: days 1 and 2, week 20 (or last dose) and follow-up day 1
- MK-8527-triphosphate (MK-8527-TP), the intracellular active metabolite, was measured in peripheral blood mononuclear cells (PBMCs) from ~20 participants/MK-8527 group at each study visit
- Pharmacokinetic exposures for MK-8527 and MK-8527-TP were based on analysis of the sparse data collected in the study

Results

Table 1. Baseline characteristics

	MK-8527 3 mg N=101	MK-8527 6 mg N=101	MK-8527 12 mg N=99	Placebo N=49
Sex at birth, n (%)				
Male	43 (42.6)	42 (41.6)	41 (41.4)	20 (40.8)
Female	58 (57.4)	59 (58.4)	58 (58.6)	29 (59.2)
Age (years), median (IQR)	27.0 (23, 33)	28.0 (24, 34)	28.0 (23, 34)	28.0 (24, 34)
Race, n (%)				
White	49 (48.5)	54 (53.5)	51 (51.5)	26 (53.1)
Black	44 (43.6)	40 (39.6)	40 (40.4)	21 (42.9)
Asian	3 (3.0)	2 (2.0)	3 (3.0)	0 (0.0)
Other	5 (5.0)	5 (5.0)	5 (5.1)	2 (4.1)
Ethnicity, n (%)				
Hispanic/Latino	18 (17.8)	13 (12.9)	14 (14.1)	11 (22.4)
Country, n (%)				
United States	42 (41.6)	48 (47.5)	45 (45.5)	21 (42.9)
South Africa	35 (34.7)	34 (33.7)	34 (34.3)	16 (32.7)
Israel	24 (23.8)	19 (18.8)	20 (20.2)	12 (24.5)

Table 2. Incidence of AEs was similar across MK-8527 and placebo groups

Number (%) of participants	MK-8527 3 mg N=101	MK-8527 6 mg N=101	MK-8527 12 mg N=99	Placebo N=49
With one or more AE	62 (61.4)	69 (68.3)	66 (66.7)	31 (63.3)
With drug-related[a] AE	15 (14.9)	16 (15.8)	20 (20.2)	9 (18.4)
With grade 3-4 AE	5 (5.0)	2 (2.0)	4 (4.0)	4 (8.2)
With SAE	2 (2.0)	0 (0.0)	1 (1.0)	1 (2.0)
With drug-related SAE	1 (1.0)	0 (0.0)	0 (0.0)	1 (2.0)
Discontinued due to an AE	0 (0.0)	2 (2.0)	1 (1.0)	2 (4.1)
Discontinued due to a drug-related AE	0 (0.0)	1 (1.0)	1 (1.0)	0 (0.0)

[a]Relationship to study drug was determined by the investigator.
One participant in the 12-mg group died of a gunshot wound (not related to study drug).
AE, adverse event; SAE, serious adverse event.

- One serious adverse event (SAE) (spontaneous abortion at ~6 weeks' gestation) was considered related to MK-8527 (3 mg)
- 2 participants discontinued MK-8527 due to drug-related AE: grade 1 CD4/lymphocyte count decreased (6 mg); grade 2 hypoesthesia (12 mg)
- No HIV-1 seroconversions were observed during the study

Table 3. Drug-related[a] AEs were similar across MK-8527 and placebo groups

Number (%) of participants	MK-8527 3 mg N=101	MK-8527 6 mg N=101	MK-8527 12 mg N=99	Placebo N=49
With drug-related[a] AE	15 (14.9)	16 (15.8)	20 (20.2)	9 (18.4)
With grade 3-4 drug-related[a] AE	1 (1.0)	0 (0.0)	1 (1.0)	1 (2.0)
Most common drug-related[a] AEs (>2% in any MK-8527 group)				
Headache	4 (4.0)	5 (5.0)	2 (2.0)	1 (2.0)
Nausea	3 (3.0)	4 (4.0)	2 (2.0)	1 (2.0)
CD4 lymphocytes decreased	2 (2.0)	1 (1.0)	3 (3.0)	1 (2.0)
Lymphocyte count decreased	0 (0.0)	1 (1.0)	4 (4.0)	1 (2.0)
Fatigue	0 (0.0)	3 (3.0)	2 (2.0)	1 (2.0)

[a]Relationship to study drug was determined by the investigator.

- Most drug-related AEs were Division of AIDS (DAIDS) toxicity grade 1 or 2 and were reported as resolved during the study
- 2 participants had confirmed CD4 and/or lymphocyte count declines that met protocol-specified discontinuation criteria, which resolved within 11 weeks
 - MK-8527 6 mg: lymphocyte nadir 0.80×10^9/L (36.5% ↓), CD4 nadir 313 cells/mm^3 (grade 1; 51.4% ↓)
 - MK-8527 12 mg: lymphocyte nadir 0.61×10^9/L (grade 1; 54.1% ↓)

Figure 2. No meaningful change in total lymphocyte count for MK-8527 doses up to 12 mg oral QM

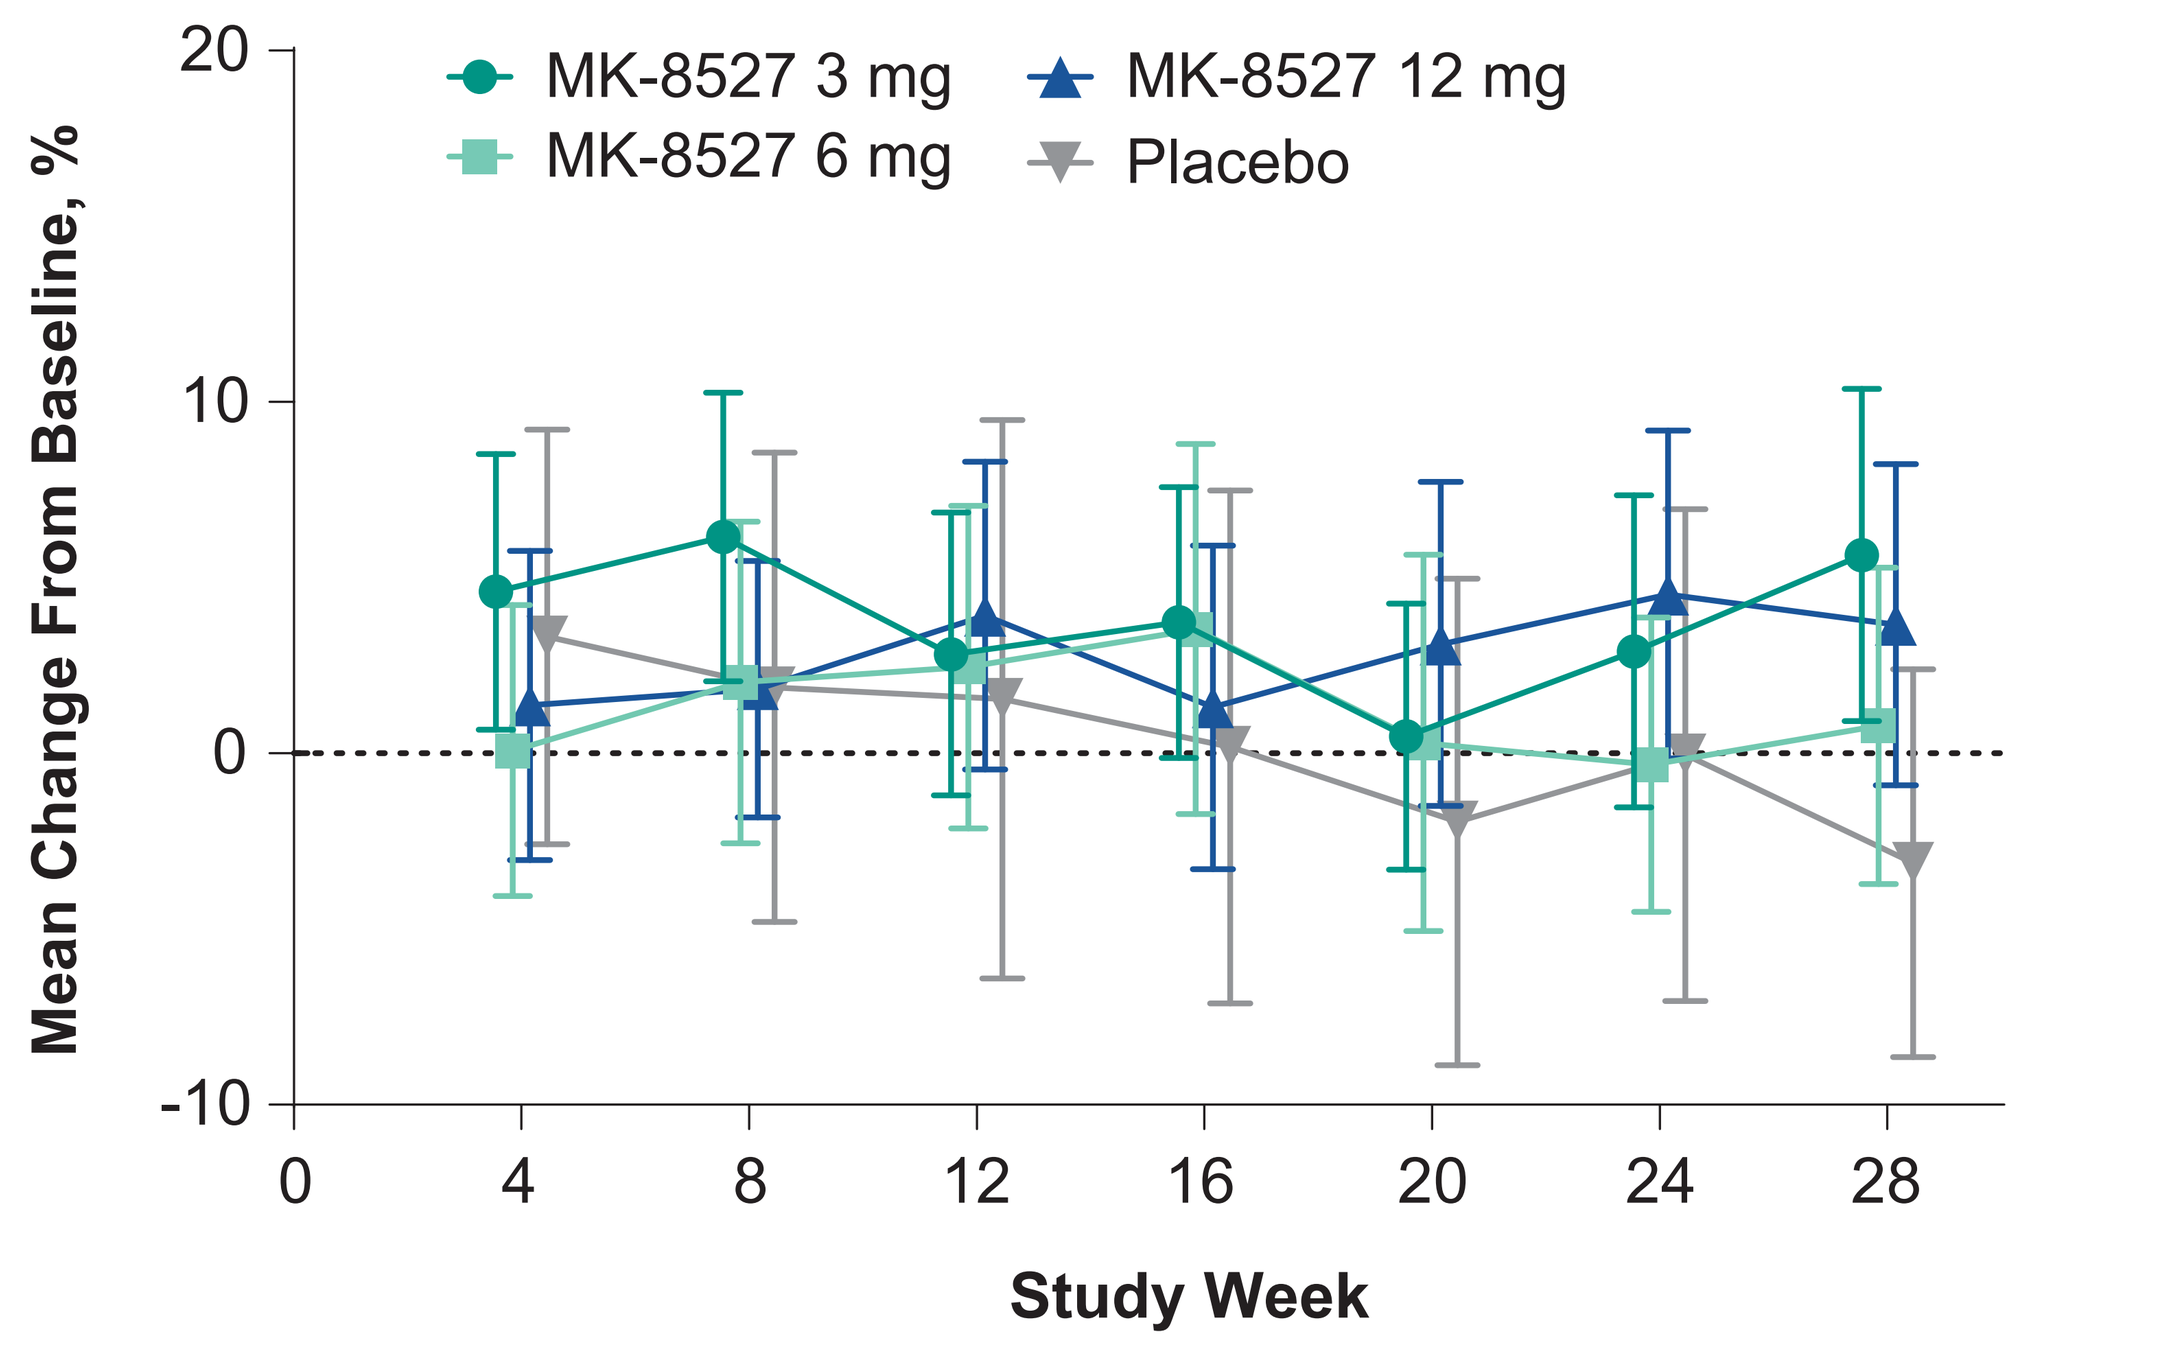
MK-8527 3 mg
MK-8527 6 mg
MK-8527 12 mg
Placebo
Mean Change From Baseline, %
Study Week

Mean % change at Week 24 (4 weeks after last dose)
3 mg: +2.90% 6 mg: -0.32% 12 mg: +4.52% Placebo: -0.04%

Figure 3. No meaningful change in CD4 count for MK-8527 doses up to 12 mg oral QM

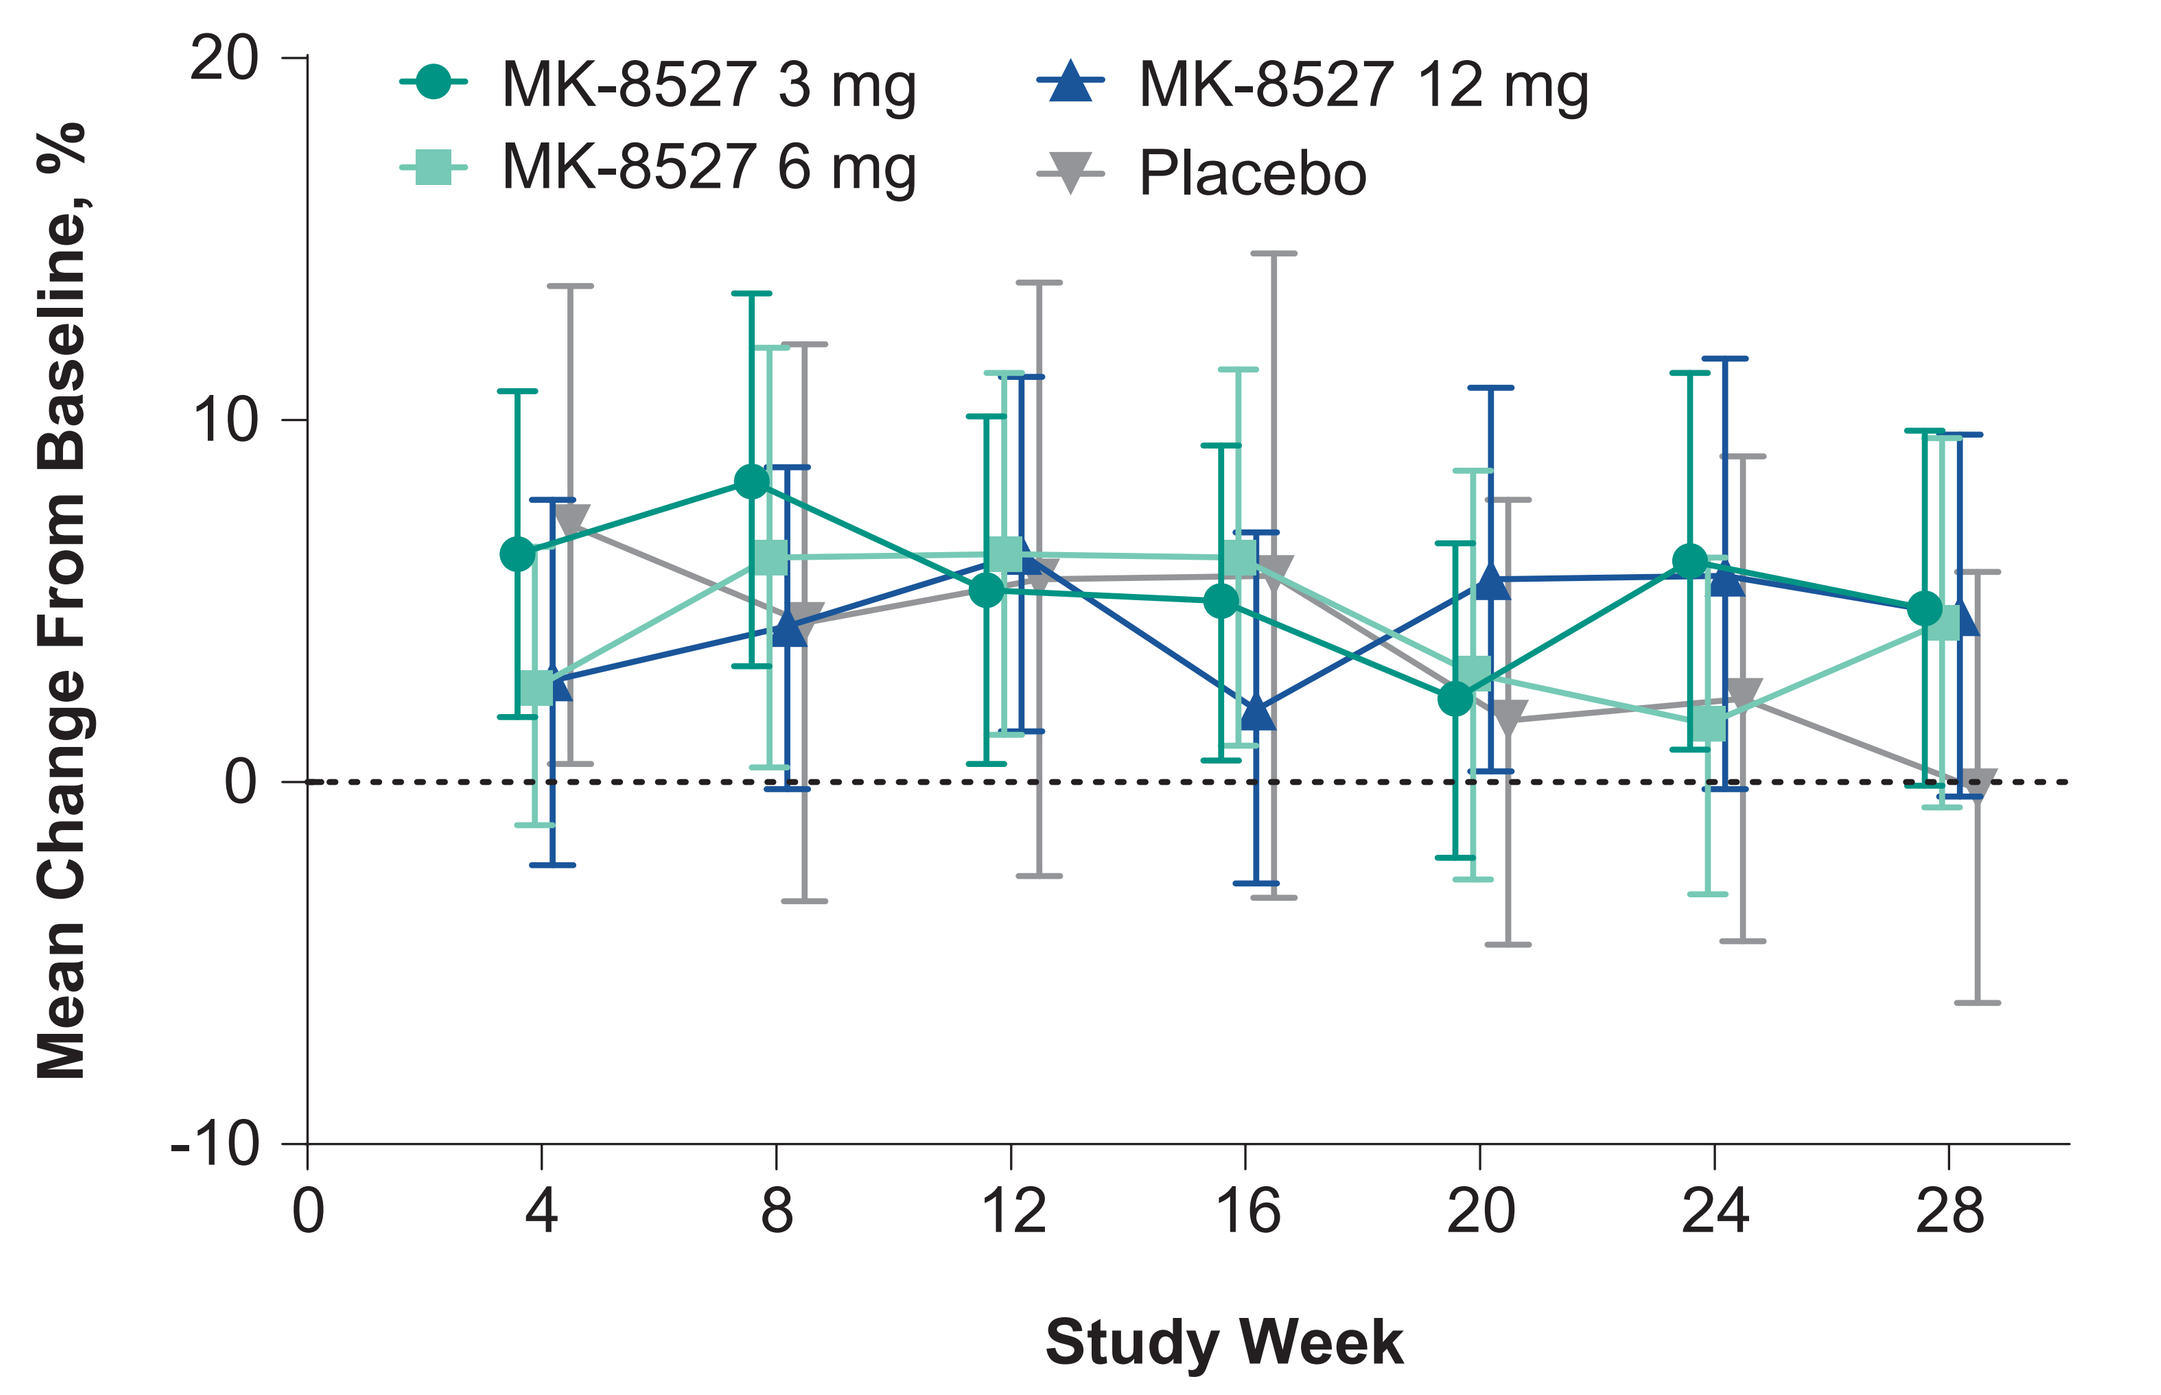
MK-8527 3 mg
MK-8527 6 mg
MK-8527 12 mg
Placebo
Mean Change From Baseline, %
Study Week

Mean % change at Week 24 (4 weeks after last dose)
3 mg: +6.1% 6 mg: +1.6% 12 mg: +5.7% Placebo: +2.3%

Figure 4. MK-8527 plasma concentration vs time profiles (semi-log scale)

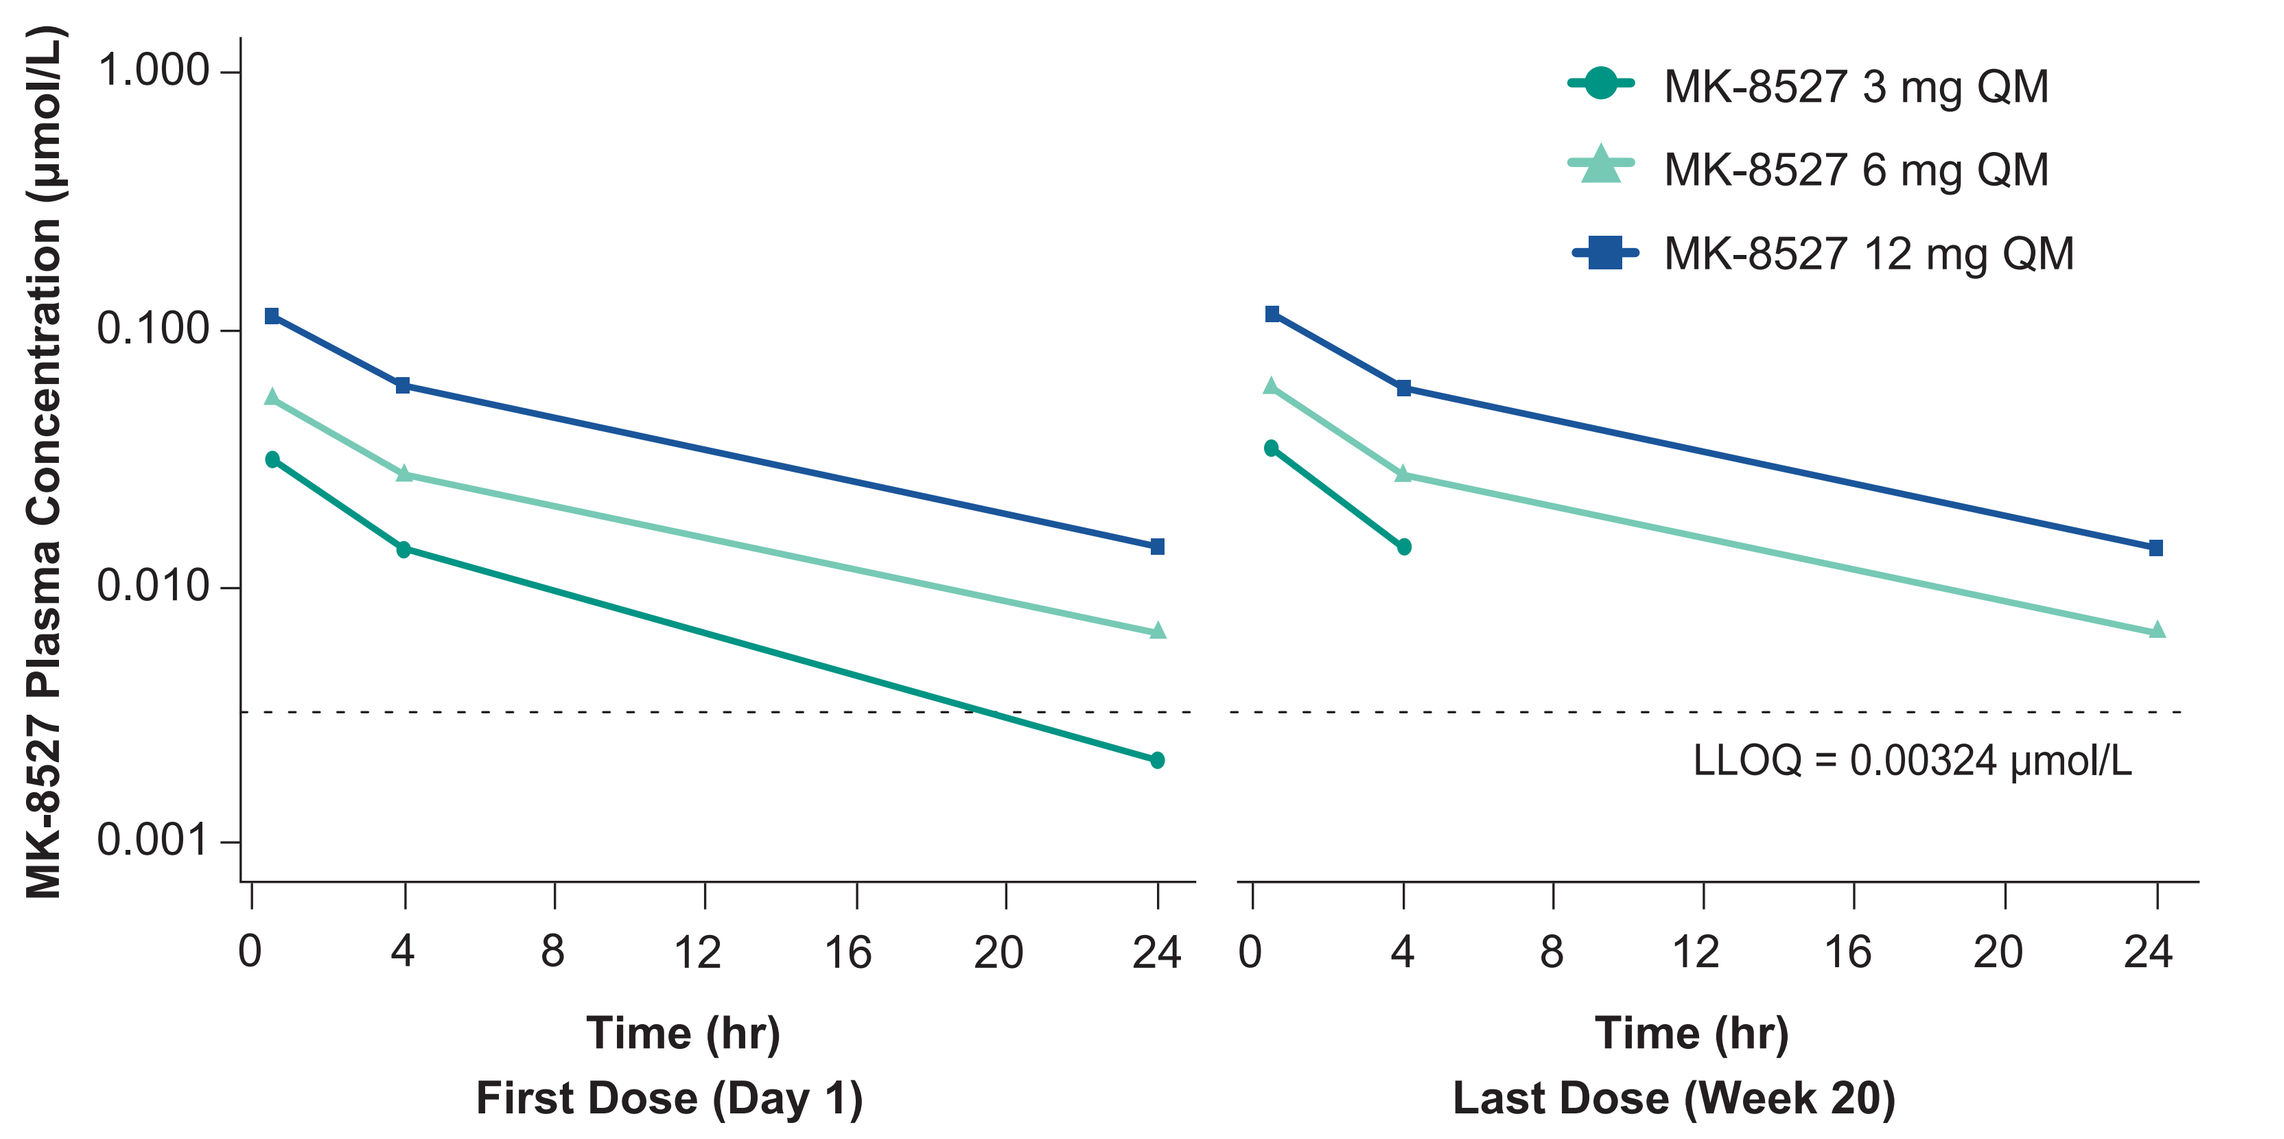
MK-8527 3 mg QM
MK-8527 6 mg QM
MK-8527 12 mg QM
MK-8527 Plasma Concentration (μmol/L)
LLOQ = 0.00324 μmol/L
Time (hr)
First Dose (Day 1)
Time (hr)
Last Dose (Week 20)

- Pharmacokinetic parameters for MK-8527 are dose-proportional
- No apparent accumulation of MK-8527 in plasma with monthly dosing

Table 4. MK-8527 Pharmacokinetic parameters

Dose, mg	Parameter	First Dose (Day 1) n	First Dose (Day 1) GM (GCV%)	Last Dose (Week 20) n	Last Dose (Week 20) GM (GCV%)
12	$AUC_{0\text{-last}}$	97	0.861 (34.1)	95	0.836 (55.0)
	C_{max}	97	0.113 (64.5)	95	0.108 (72.5)
6	$AUC_{0\text{-last}}$	96	0.397 (36.1)	92	0.404 (37.3)
	C_{max}	96	0.0490 (75.6)	92	0.0538 (73.5)
3	$AUC_{0\text{-last}}$	95	0.132 (84.8)	78	0.129 (81.5)
	C_{max}	95	0.0289 (69.0)	78	0.0320 (67.5)

AUC, area under the curve (hr*μmol/L). C_{max}, maximum concentration in plasma (μmol/L); GM, geometric mean. GCV%, geometric coefficient of variation.

Figure 5. MK-8527-TP PBMC concentration vs time profiles (semi-log scale)

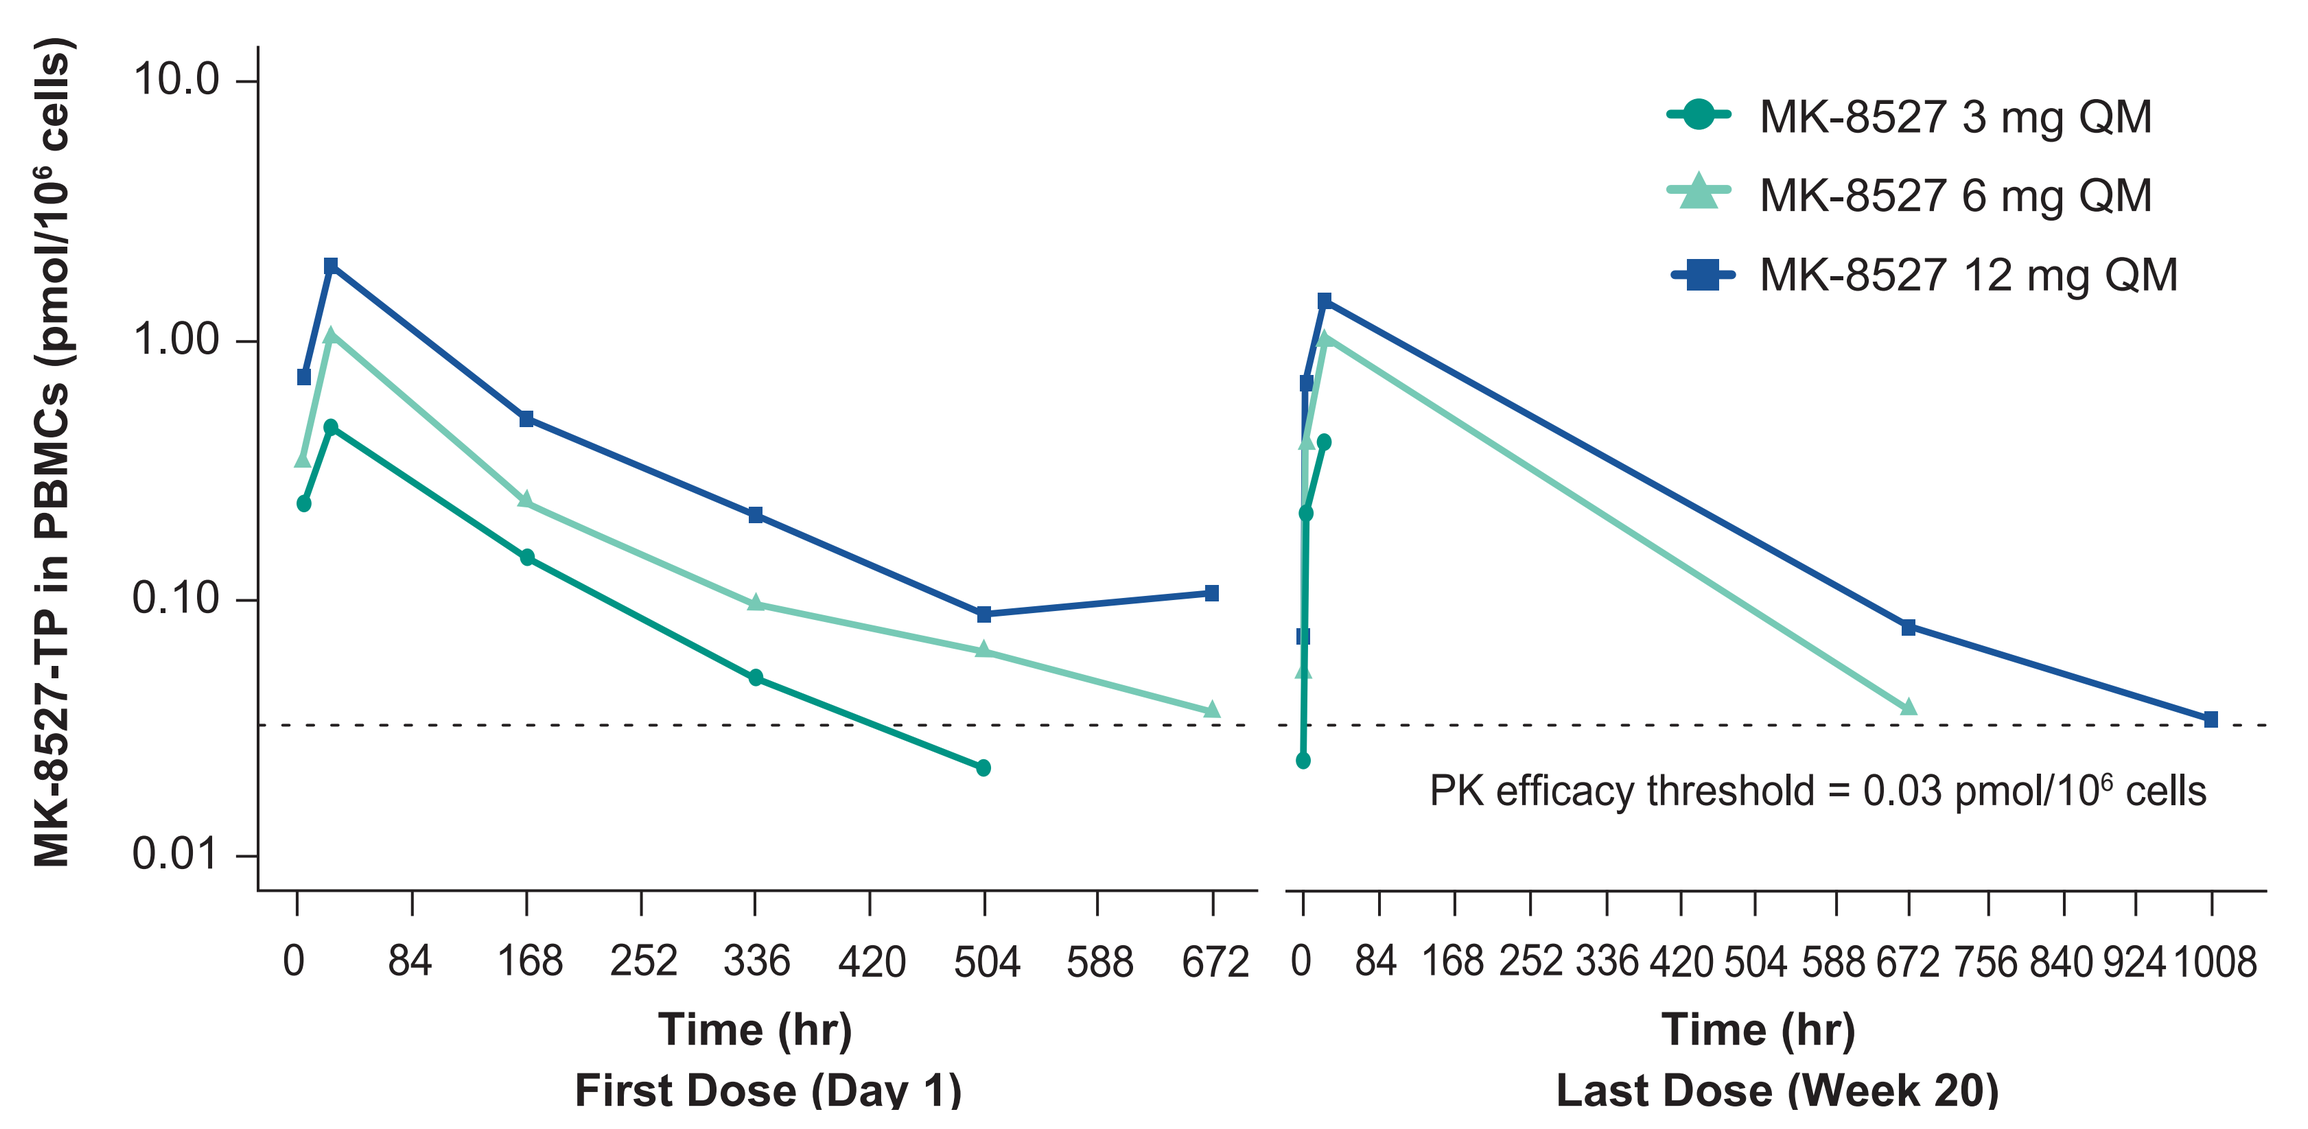
MK-8527 3 mg QM
MK-8527 6 mg QM
MK-8527 12 mg QM
MK-8527-TP in PBMCs (pmol/10⁶ cells)
PK efficacy threshold = 0.03 pmol/10⁶ cells
Time (hr)
First Dose (Day 1)
Time (hr)
Last Dose (Week 20)

- Pharmacokinetic parameters for MK-8527-TP are dose-proportional
- Minimal accumulation of MK-8527-TP in PBMCs with monthly dosing

Table 5. MK-8527-TP Pharmacokinetic parameters

Dose, mg	Parameter	First Dose (Day 1) n	First Dose (Day 1) GM (GCV%)	Last Dose (Week 20) n	Last Dose (Week 20) GM (GCV%)
12	$AUC_{0\text{-last}}$	22	204 (71.3)	19	287 (55.9)
	C_{max}	22	1.48 (79.7)	19	1.23 (63.9)
6	$AUC_{0\text{-last}}$	26	108 (56.3)	23	128 (178.6)
	C_{max}	26	0.707 (89.7)	23	0.782 (72.2)
3	$AUC_{0\text{-last}}$	21	52.9 (67.7)	20	20.7 (255.5)
	C_{max}	21	0.394 (89.4)	20	0.325 (78.7)

AUC, area under the curve (hr*pmol/10^6 cells). C_{max}, maximum concentration in PBMC (pmol/10^6 cells). GM, geometric mean. GCV%, geometric coefficient of variation.

Conclusions

- In this large, randomized, double-blind, placebo-controlled, Phase 2 study in adults with low likelihood of HIV-1 exposure:
 - 6 consecutive monthly doses of MK-8527 (3 mg, 6 mg, and 12 mg) were generally well tolerated and demonstrated a safety profile similar to placebo
 - Pharmacokinetics of MK-8527 and MK-8527-TP were dose-proportional and well-correlated
- These results support the continued clinical development of MK-8527 oral QM for PrEP
- Phase 3 studies of MK-8527 oral QM, EXPrESSIVE-10 and EXPrESSIVE-11, are currently enrolling participants

MK-8527 phase 3 program for PrEP

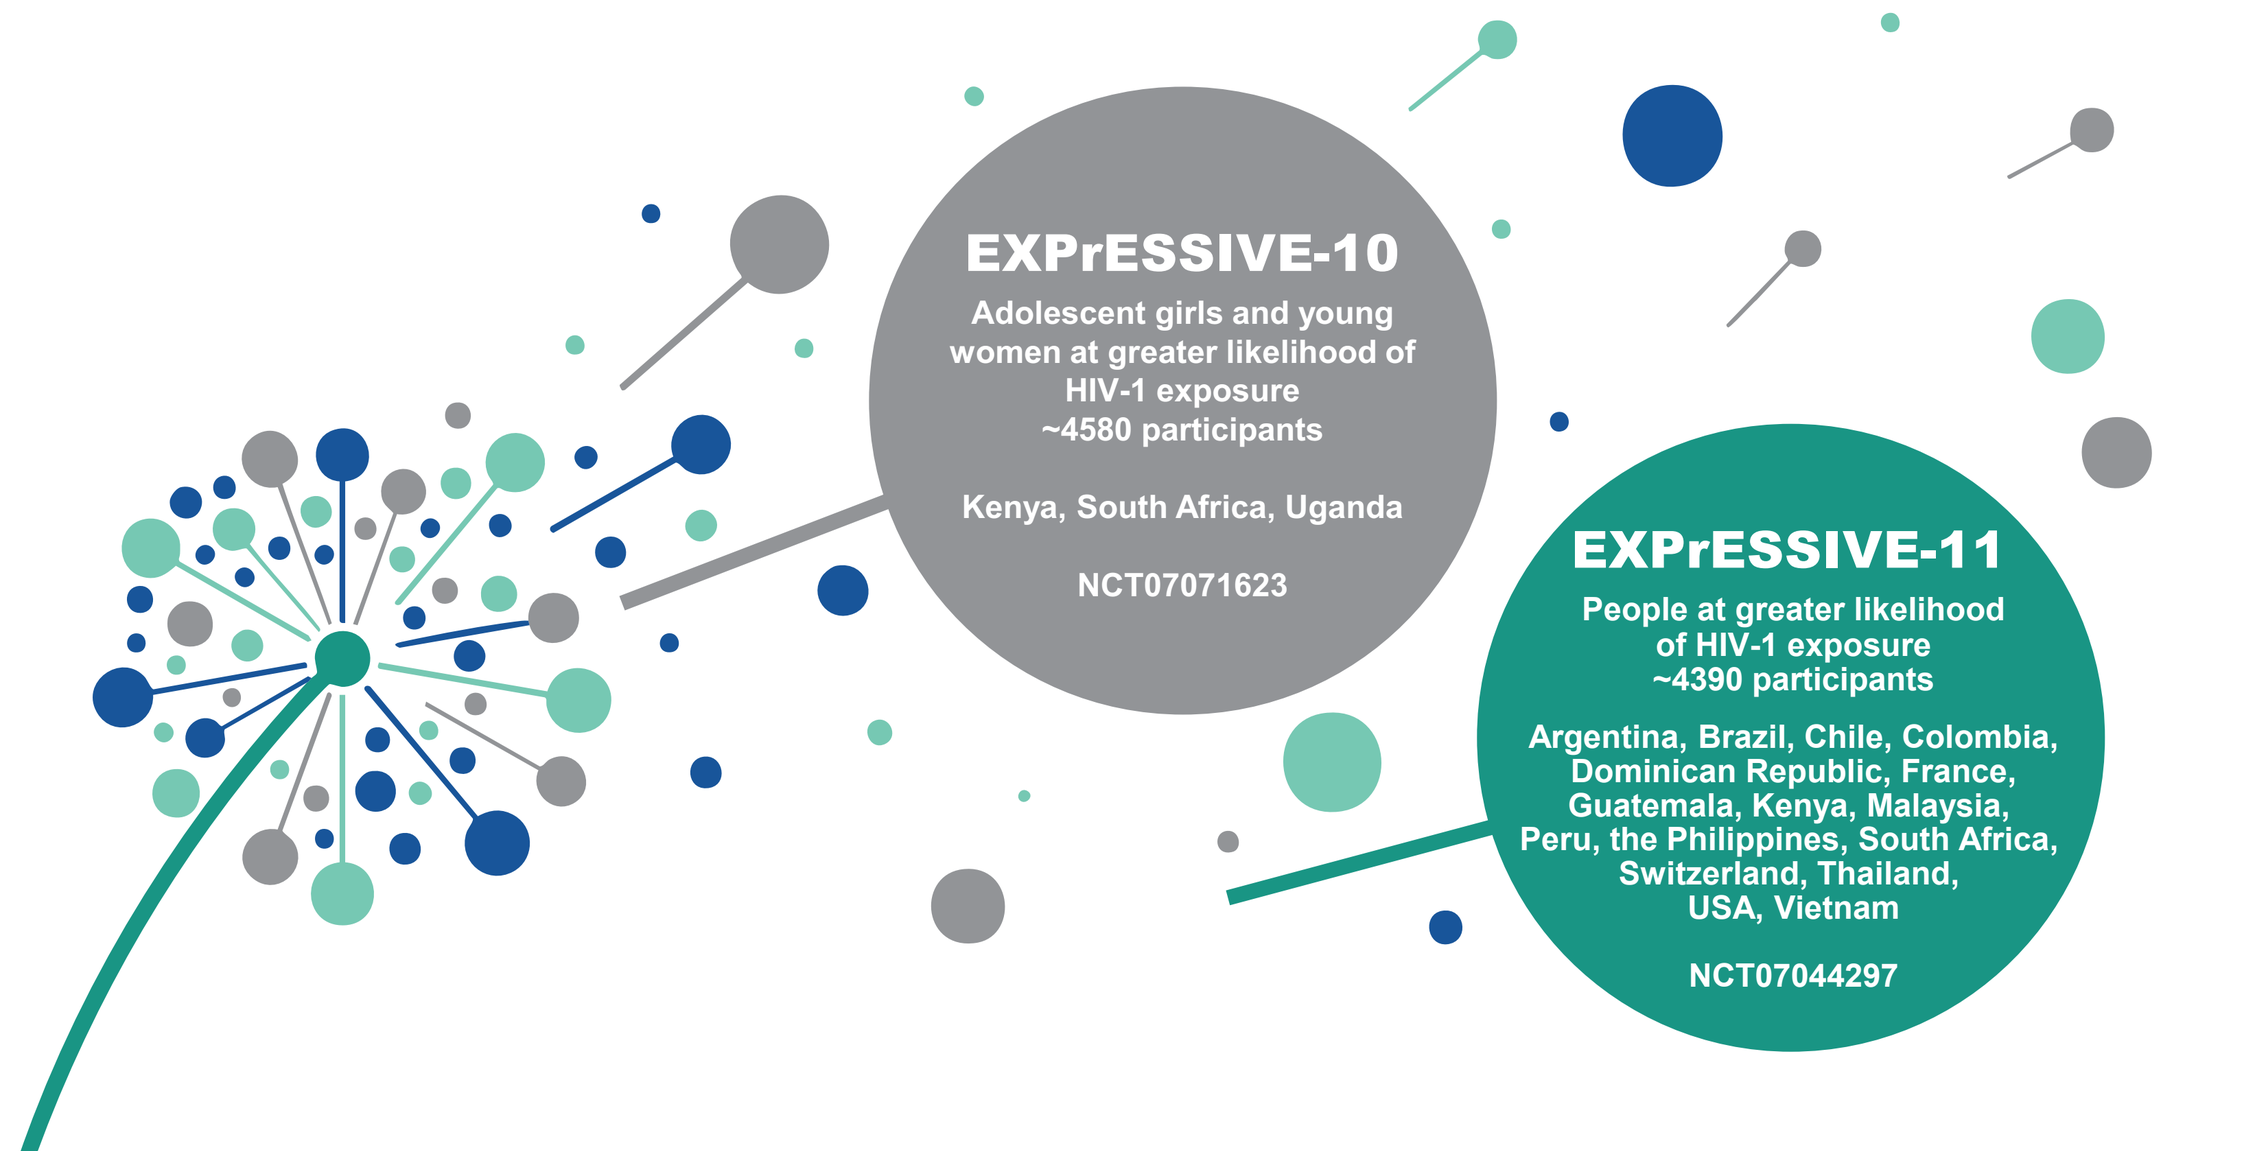
EXPrESSIVE-10
Adolescent girls and young women at greater likelihood of HIV-1 exposure
~4580 participants
Kenya, South Africa, Uganda
NCT07071623
EXPrESSIVE-11
People at greater likelihood of HIV-1 exposure
~4390 participants
Argentina, Brazil, Chile, Colombia, Dominican Republic, France, Guatemala, Kenya, Malaysia, Peru, the Philippines, South Africa, Switzerland, Thailand, USA, Vietnam
NCT07044297

References

1. WHO, HIV data and statistics, available at: https://www.who.int/teams/global-hiv-hepatitis-and-stis-programmes/hiv/strategic-information/hiv-data-and-statistics accessed June 12, 2025.
2. O Murchu E, et al. *BMJ Open.* 2022;12:e048478
3. Hojilla JC, et al. *JAMA Network Open.* 2021;4(8):e2122692
4. Gravett RM, et al. *J Acquir Immune Defic Syndr.* 2025;99:128-137.
5. Gillespie G, et al. Safety and Pharmacokinetics of MK-8527, a Novel NRTTI, in Adults without HIV. Presented at 31st CROI, March 3-6, 2024, Denver CO.

Acknowledgments

We thank the participants and their families and the investigators and site personnel from 18 study sites.

Contact Information

Contact Boris Renjifo at boris.renjifo@merck.com for questions or comments.

Disclosures

The presenting author (Boris Renjifo) declares the following potential conflicts of interest: Employee of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, who may own stock and/or hold stock options in Merck & Co., Inc., Rahway, NJ, USA.
Funding for this research was provided by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.
Employees of the study sponsor were involved in trial design, data analysis, and co-authoring this presentation.
Medical writing assistance was provided by Kim Strohmaier, MPH, and editorial assistance by Carol Zecca, BS, both of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

To download a plain language summary
https://bit.ly/43SGlrY

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the Congress or the author of this poster.

To download this poster
https://bit.ly/4ae5WjT

Presented at South Atlantic Association of Obstetricians and Gynecologists (SAAOG) Annual Meeting; White Sulphur Springs, WV, USA; January 24-26, 2026.

Copyright © 2025 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.